Three Year Follow-up in an Open-Label, Pilot Study of Transendocardial Autologous Bone Marrow in Chronic Myocardial Infarction using a Helical Needle Catheter

Luis M. de la Fuente, M.D.*, Simon H. Stertzer, M,D. ¥, Eduardo Peñaloza, M.D.*, Julio Argentieri, M.D.* , Jorge Miano, M.D.* , Benjamin Koziner, M.D.*,
Cristian Bilos M.D.*, Licia Battaglia, Ph.D.[#], Graciela Lucero Ph.D.[#], Jessica Stertzer[§], Didier Rouy, M.D. Ph.D.[§], Peter A. Altman, PhD[§]

***Instituto Argentino del Diagnostico y Tratamiento and Clinica Suiza, Buenos Aires, Argentina, [¥]Stanford University Medical Center, Palo Alto, CA, [#]Cell Prep SA, Buenos Aires, Argentina, [§]BioCardia Inc., South San Francisco, CA**

Abstract

Background: We assessed the hypothesis that helical needle transendocardial delivery of autologous bone marrow (ABM) mononuclear cells around regions of hypo or akinesia in chronic myocardial infarction (MI) patients would be safe and possibly improve ejection fraction (EF).

Methods: 20 stable post-MI ischemic heart failure patients with an EF <40% were enrolled. ABM cells were aspirated from the iliac crest, washed, filtered, counted for CD34+ and CD133+ content, resuspended at a concentration of 100 million cells/cc, and delivered to the heart percutaneously from 5 to 10 approximate peri-infarct sites with a transendocardial helical needle catheter and steerable guide catheter platform. 2D echo left ventricle EF measurements, 24 hour Holter, and exercise tolerance testing were performed at baseline, day of procedure, 1 and 12 weeks, 6, 12 and 24 months. An additional follow-up was performed at 36 months.

Results: An average of 96 million ABM mononuclear cells were injected into 8.5±2.6 sites around the infarct to target the peri-infarct zones (n=20). There were no adverse events associated with the catheter based cell transplantation procedure in any of the 20 treated patients. There has been no significant increase in the averaged number of ventricular events on 24h Holter. In patients with 3 year follow-up (n=10), EF significantly increased over baseline (35.2±4.6 %) at each time point: 6 months (40.8±4.5, p=0.003), 12 months (42.3±5.1, p=0.0001), 24 months (42.3 ±6.1, p = 0.0008) and 36 months (43.7 ±7.3, p = 0.0001). The average end diastolic ventricular dimension and exercise tolerance trended toward benefit, but differences were not statistically significantly.

Conclusions: ABM cells delivered with the helical needle transendocardial catheter was safe in this uncontrolled study in patients with chronic MI. Increased ejection fraction and other positive data trends support the ongoing randomized trial of this therapeutic strategy.

Test or procedure	Baseline	Trans-plant	Follow-up visit windows: Weekly visits (±3 days)		Monthly visits (±7 days)			
			W1	W12	M6	M12	M24	M36
ABM Aspiration		X						
Cell Implantation		X						
Physical Exam	X		X	X	X	X	X	X
ECG	X		X	X	X	X	X	X
Echo	X	X		X	X	X	X	X
24 Hour Holter	X		X	X	X	X	X	
Exercice Tolerance Testing	X			X	X	X	X	
Blood Tests	X	X	X	X	X	X	X	
Urinalysis	X		X	X	X	X	X	
Adverse Events	X	X	X	X	X	X	X	X
Concomitant Meds	X	X	X	X	X	X	X	X

Figure 1. Delivery through helical needle catheter from within the heart

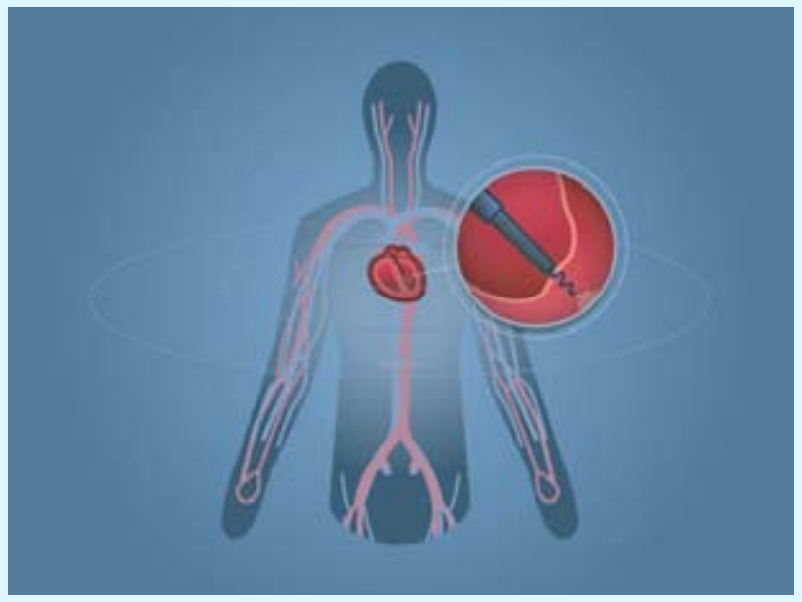

Figure 2. The Helix® catheter for transendocardial delivery is advanced through a steerable guide (not shown). In addition to the therapeutic lumen which discharges at the distal tip of the helix, it has a second lumen which discharges from the base of the helix for delivering contrast to confirm engagement and positioning.

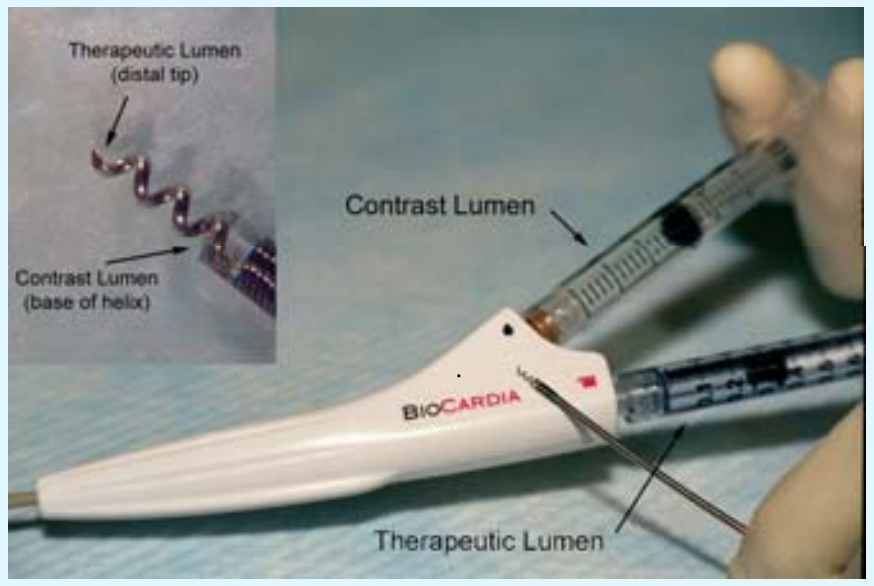

Therapeutic Lumen (distal tip)
Contrast Lumen (base of helix)
Contrast Lumen
BIOCARDIA
Therapeutic Lumen

Figure 3. Ejection fraction over time for both groups of patients

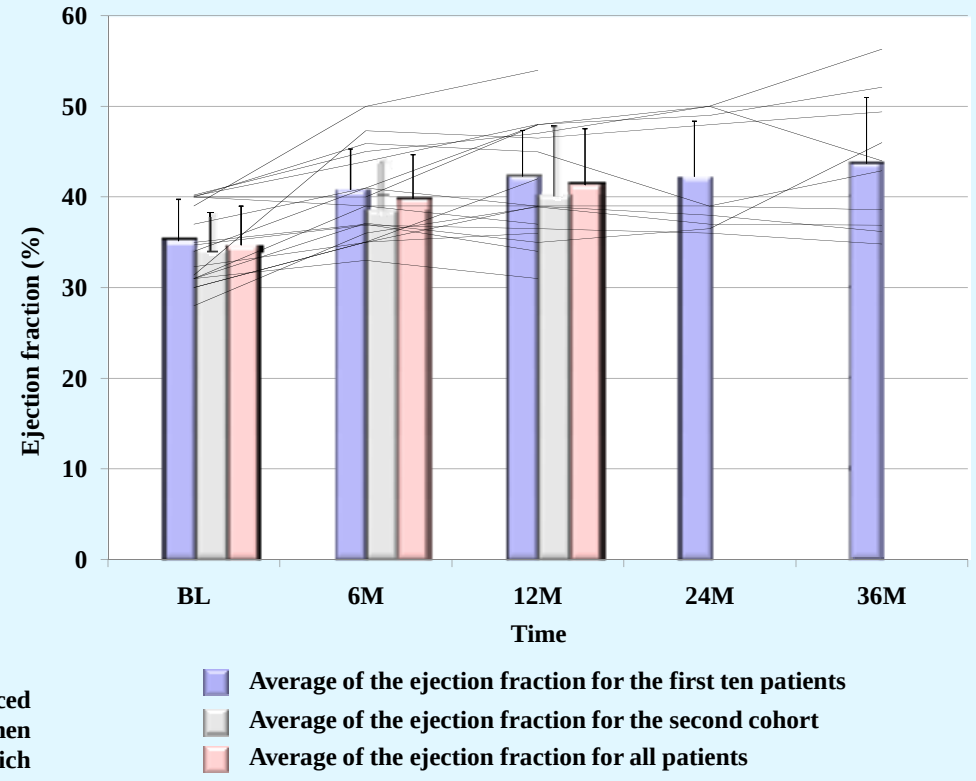

Ejection fraction (%)
60
50
40
30
20
10
0
BL
6M
12M
24M
36M
Time
Average of the ejection fraction for the first ten patients
Average of the ejection fraction for the second cohort
Average of the ejection fraction for all patients

Figure 4. Changes in Exercise Tolerance Time for eighteen patients. Patient #11 died in elective heart transplant just before 6 month follow-up, and patient #20 is approaching one year follow-up. Of note, some patients have the same values, and are shown as only one line.

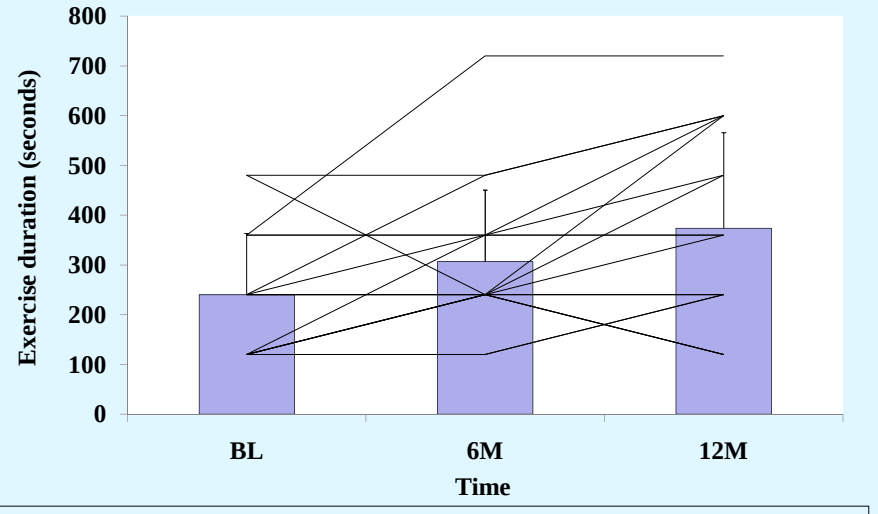

Exercise duration (seconds)
800
700
600
500
400
300
200
100
0
BL
6M
12M
Time

Methods

The one year results of the first cohort of the present study was published in the American Heart Journal (de la Fuente LM, Stertzer SH, Argentieri J et al. Transendocardial autologous bone marrow in chronic myocardial infarction using a helical needle catheter: 1-year follow-up in an open-label, nonrandomized, single-center pilot study (the TABMMI study). *Am Heart J 2007 July;154(1):79-7.* , where the methods are fully described. The second cohort of the study can be summarized as follows.

<u>Population Studied.</u> Patients were enrolled who had sustained a transmural myocardial infarction and showed echocardiographic evidence of left ventricular dysfunction having ejection fractions of <40%.

<u>Marrow Harvest and Processing.</u> 50 cc of autologous bone marrow cells were collected under local anesthesia from the posterior iliac crest. Bone marrow mononuclear cells were isolated by density gradient on Ficoll-Paque Plus tubes and resuspended in Ringers solution at a concentration of 1 x 108 cells/ml in a total volume of 1.3 ml.

<u>Catheter Delivery of Cells.</u> Upon catheterization, the selected target regions were assessed using fluoroscopy. Three investigators established anatomic consensus regarding the helical needle placement in predefined target regions. Cells were delivered transendocardially with the two-catheter Helical Infusion System for transendocardial delivery (BioCardia, Inc., South San Francisco, CA).

<u>First cohort follow-up.</u> The ten first patients were followed for two years. An additional one year follow up was also performed which was not specified in the protocol. The ejection fraction is presented figure 3.

<u>Second cohort follow-up.</u> The ten patients included in the second cohort were followed for one year with the same endpoints as the first cohort.

Results and discussion

In this open labelled safety study, twenty patients were enrolled in two cohorts. One-year data on the first cohort of ten patients has been previously published (de la Fuente et al. American Heart Journal 2007).

<u>First cohort: patients 1 to 10</u>
Three year follow-up was recently completed although the protocol required only two. Three year echocardiography was performed.
<u>Second cohort: patients 11 to 20</u>
Ten more patients were enrolled, nine of which have reached their one year follow-up. One patient died during an elective heart transplant 177 days after the procedure. This patient had been on a heart transplant list before being enrolled, and a donor heart became available. No other serious adverse event was noticed.

Figure 3 shows the left ventricular ejection fraction measured by transthoracic 2D echo for all twenty patients. In the first cohort, the resulting averaged ejection fraction remains statistically improved over baseline at 24 months (p=0.0008) and 36 months (p=0.0001). In the second cohort, six-month data is also improved over baseline.

Figure 4 shows the exercice tolerance time for 18 of the 20 patients enrolled. Analysis at one year is statistically significant (p=0.008 at 12 months versus baseline) but needs to be confirmed with the last patient's one year follow-up.

For the first cohort, no adverse event was reported between two and three years for any of these ten patients. For the second cohort, no serious adverse event was reported in addition to the death during the elective transplant.

The transthoracic echo measurements of ejection fraction demonstrate an average improvement at 36 months in this pilot study. However, it is noted that one patient had a precipitous fall in ejection fraction from baseline to 3 months and that this significant change reversed itself. These unpredicted improvements in EF at 36 months are not powered to confirm the clinical efficacy of this therapy but they are compelling in a safety pilot trial.

Helical needle transendocardial delivery provided the investigators with a controlled and stable delivery means to target segmental dyskinesia using fluoroscopy. The helical fixation needle may reduce back-leak into the ventricular chamber by providing a simplified direct means to confirm that one is positioned within the myocardium, a longer self sealing helical pathway into the tissue, and the ability to leave the helix in place after delivery to act as a barrier to obstruct back-leak, and control the time course of delivery

<u>Conclusions:</u>

In this open labeled uncontrolled safety trial of helical needle transendocardial delivery of autologous bone marrow, the lack of procedural complications, the statistically significant improvements in LV ejection fraction and exercise tolerance support the safety of this therapy and support the ongoing randomized trial of this therapeutic strategy.